UBX1325, a small molecule inhibitor of Bcl-xL, attenuates vascular dysfunction in two animal models of retinopathy

Pamela Tsuruda[1], Shawnta Chaney[1], Agnieszka Dejda[2], Sonali Dasgupta[1], Sergio Crespo-Garcia[2], Surabi Rao[1], Scott Armstrong[1], Dan Marquess[1], Przemyslaw Sapieha[2], Pedro Beltran[1]

[1]UNITY Biotechnology, South San Francisco (CA), USA; [2]Maisonneuve-Rosemont Hospital Research Centre, University of Montreal, Montreal (QC), Canada

Introduction

Retinal vasculopathies account for the primary causes of loss of sight in the industrialized world and current standards do not fully address these diseases. Focused on novel treatment paradigms, we developed UBX1325, a novel small molecule inhibitor prodrug of specific subtypes within the B-cell lymphoma 2 (Bcl-2) family of apoptosis regulatory proteins and assessed its efficacy in senescence-associated models of retinopathy[1].

Methods

- Target engagement (TE), decreased Bcl-xL:Bim or Bcl-2:Bim complexes, was measured by an electro-chemiluminescence-based assay in cell or retinal lysates from C57BL/6 mice after intravitreal (IVT) injection of UBX1325.
- Senolysis was measured as cell viability (CellTiter-Glo) after 72h incubation of senescent HRMEC with the UBX1325 parent.
- Initiation of apoptosis (mechanism engagement, ME) was measured by caspase-3/7 activation.
- Adult mice and neonatal mice from oxygen induced retinopathy (OIR; 75% O_2 from post-natal day (P)7-P12) or normoxic controls were used for TE and ME after IVT injection of UBX1325 at P12.
- Vascular endpoints (neovascular and avascular area) were evaluated at P17 by isolectin B4 staining in OIR mice given a single IVT injection of UBX1325 at P12.
- UBX1325 was studied in the streptozotocin (STZ)-induced retinopathy model. UBX1325 was injected IVT at weeks 8 and 9 post-STZ, and retinal endpoints were measured at week 10. Vascular leakage was evaluated by Evans blue dye extravasation into the retina after intravenous injection. The dark-adapted electroretinogram was used to assess retinal function with increasing flash intensity.

Results

UBX1325 is a Bcl-xL inhibitor prodrug

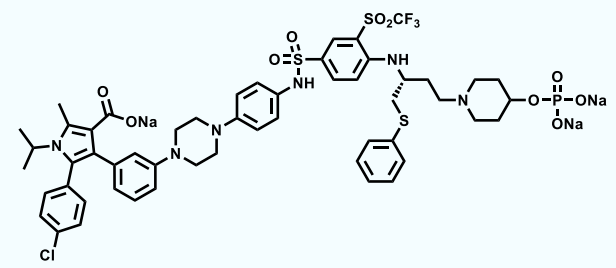
SO2CF3
NH
ONa
O
Cl
N
S
O-P-ONa
ONa

	HRMEC TE (pIC_{50}) (95% CI)	HRMEC ME (pEC_{50}) (95% CI)	HRMEC senolysis (pEC_{50}) (± SD)
UBX1325 parent	**8.1** (7.8, 8.6)	**7.3** (∞, 8.2)	**7.8** (±0.6)

IVT UBX1325 leads to a reduction of anti-apoptotic Bcl-xL:Bim complexes in the adult mouse retina

- IVT injection of UBX1325 results in selective inhibition of Bcl-xL in the mouse retina
- A single IVT administration of UBX1325 results in dose- and time-dependent inhibition of Bcl-xL
- Bcl-xL inhibition does not induce apoptosis in a healthy adult mouse retina

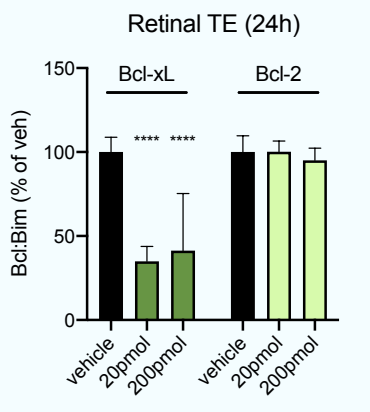
Retinal TE (24h)
Bcl-xL
Bcl-2
Bcl:Bim (% of veh)
vehicle
20pmol
200pmol

****p<0.0001 v. vehicle by two-way ANOVA with Dunnett's multiple comparisons test

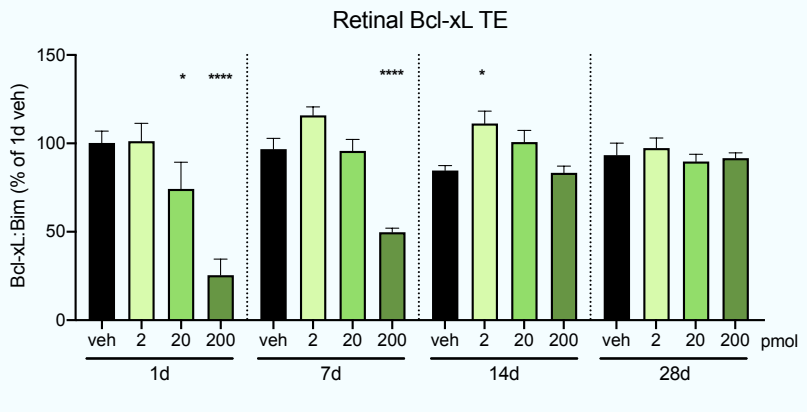
Retinal Bcl-xL TE
Bcl-xL:Bim (% of 1d veh)
veh 2 20 200 pmol
1d
7d
14d
28d

*p<0.05, ****p<0.0001 v. vehicle by two-way ANOVA with Dunnett's multiple comparisions test

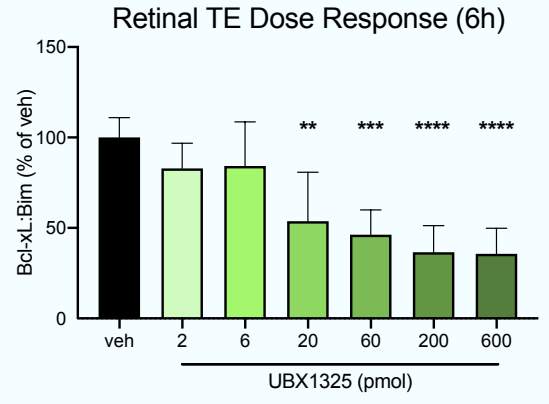
Retinal TE Dose Response (6h)
Bcl-xL:Bim (% of veh)
veh
2
6
20
60
200
600
UBX1325 (pmol)

p<0.01, *p<0.001, ****p<0.0001 v. vehicle by one-way ANOVA with Dunnett's multiple comparisions test

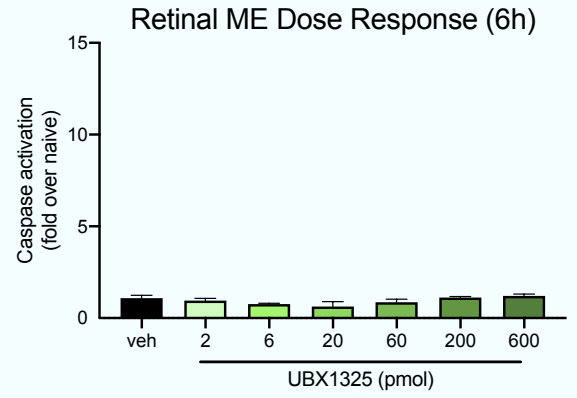
Retinal ME Dose Response (6h)
Caspase activation (fold over naive)
veh
2
6
20
60
200
600
UBX1325 (pmol)

IVT UBX1325 promotes apoptosis in OIR, but not normoxic mice

- IVT injection of UBX1325 results in 24h Bcl-xL TE (37-81%) in OIR and normoxic animals
- Selective caspase-3/7 activation was observed (3-9-fold) in OIR animals 24h after IVT administration of UBX1325

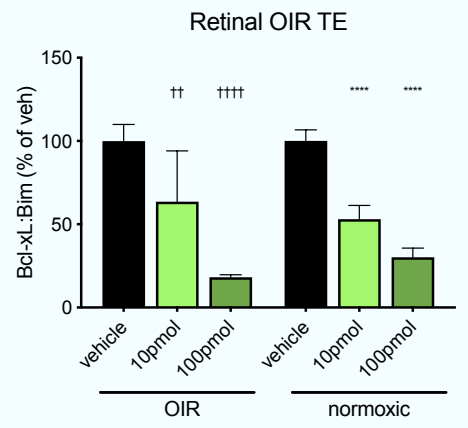
Retinal OIR TE
Bcl-xL:Bim (% of veh)
vehicle
10pmol
100pmol
OIR
normoxic

††p<0.01; ††††p<0.0001 v. OIR vehicle; ****p<0.0001 v. normoxic vehicle by one-way ANOVA with Dunnett's multiple comparisons test

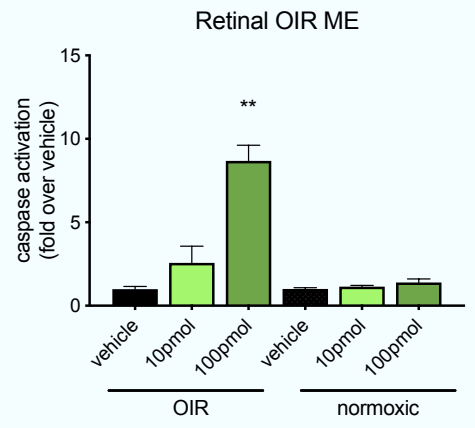
Retinal OIR ME
caspase activation (fold over vehicle)
vehicle
10pmol
100pmol
OIR
normoxic

**p<0.01 v. OIR vehicle by one-way ANOVA with Dunnett's multiple comparisons test

UBX1325 improves retinal vascular phenotypes in the mouse OIR model

- IVT injection of UBX1325 improves both retinal neovascularization (58-71%) and avascular area (32-52%)

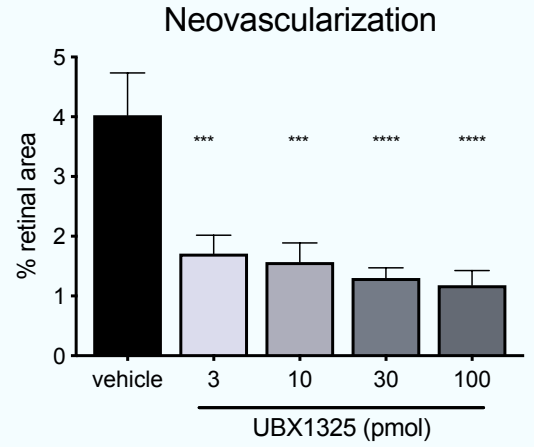
Neovascularization
% retinal area
vehicle
3
10
30
100
UBX1325 (pmol)

*** p<0.001, **** p<0.0001 v. vehicle by one-way ANOVA, with Dunnett's multiple comparisons test

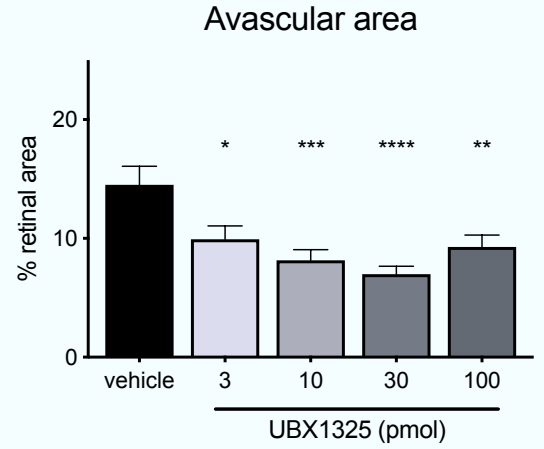
Avascular area
% retinal area
vehicle
3
10
30
100
UBX1325 (pmol)

* p<0.05; ** p<0.01; *** p<0.001, **** p<0.0001 v. vehicle by one-way ANOVA, with Dunnett's multiple comparisons test

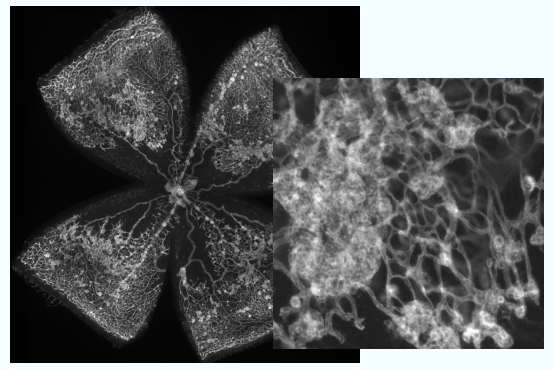

vehicle

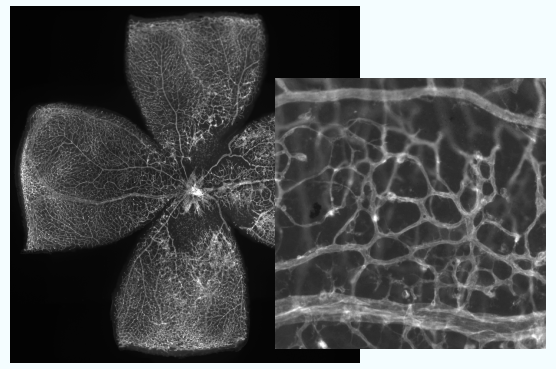

30pmol UBX1325

UBX1325 reduces vascular leakage and improves retinal function in the diabetic mouse

- IVT injection of UBX1325 reduced retinal vascular permeability (78-90%) and improved ERG (a- and b-wave amplitude)

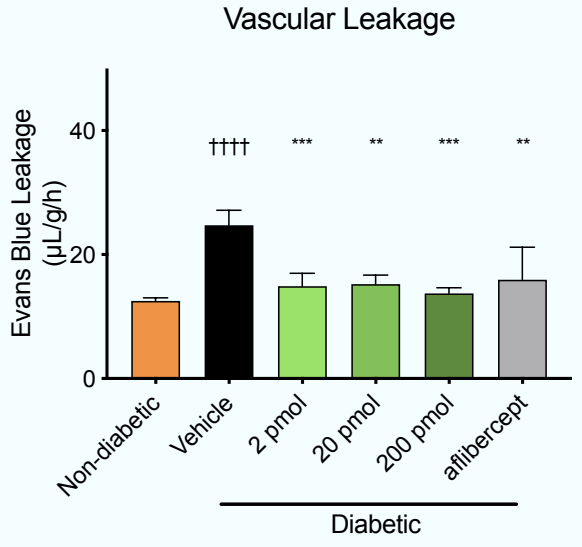
Vascular Leakage
Evans Blue Leakage (µL/g/h)
Non-diabetic
Vehicle
2 pmol
20 pmol
200 pmol
aflibercept
Diabetic

†††† p<0.0001 v. Non-diabetic control by two-tailed t-test
p<0.01; *p<0.001 v. Vehicle by one-way ANOVA with Dunnett's multiple comparisons test

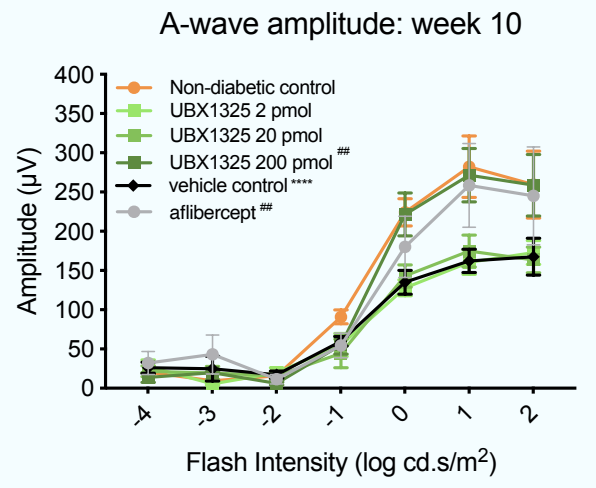
A-wave amplitude: week 10
Non-diabetic control
UBX1325 2 pmol
UBX1325 20 pmol
UBX1325 200 pmol ##
vehicle control ****
aflibercept ##
Amplitude (µV)
Flash Intensity (log cd.s/m2)

**** p<0.0001 v. Non-diabetic control by two-way ANOVA
p<0.01 v. Vehicle control by two-way ANOVA with Dunnett's multiple comparisons test

Conclusions

Inhibition of retinal Bcl-xL by UBX1325 promotes apoptosis in the senescence-associated OIR model. Although anti-VEGF agents decrease neovascularization in OIR[2], UBX1325 is differentiated as it improves both the retinal neovascular and avascular areas in the OIR model. UBX1325 also improves vascular leak and ERG in STZ mice. Collectively, our data support development of UBX1325 for retinal vasculopathies and initial clinical evaluation in patients with diabetic macular edema (DME)[3].

[1]**Pathological angiogenesis in retinopathy engages cellular senescence and is amenable to therapeutic elimination via BCL-xL inhibition** (2021). S Crespo-Garcia, PR Tsuruda, A Dejda, RD Ryan, F Fournier, SY Chaney, F Pilon, T Dogan, G Cagnone, P Patel, M Buscarlet, S Dasgupta, G Girouard, SR Rao, AM Wilson, R O'Brien, R Juneau, V Guber, A Dubrac, C Beausejour, S Armstrong, FA Mallette, CB Yohn, J-S Joyal, D Marques, PJ Beltran, P Sapieha. 33(4): 818-832

[2]**Senescence-associated secretory phenotype contributes to pathological angiogenesis in retinopathy** (2016). M Oubaha, K Miloudi, A Dejda, V Guber, G Mawambo, M-A Germain, G Bourdel, N Popovic, FA Rezende, RJ Kaufman, FA Mallette, P Sapieha. 8(362): 362ra144

[3]**Safety and Tolerability Study of UBX1325 in Patients With Diabetic Macular Edema or Neovascular Age-Related Macular Degeneration.** UNITY Biotechnology. NCT04537884